Survival CSF Collection: A novel method for serial collection of cerebrospinal fluid from rats

Dany Gitnick, Edwin Custodio, and J. Eric McDuffie Investigative Toxicology, Neurocrine Biosciences, Inc., San Diego, CA USA 92130

ABSTRACT

The Blood Brain Barrier is a specialized cellular barrier critical to controlling the passage of substances into the cerebrospinal fluid (CSF), which protects the brain against circulating toxins and pathogens. Many central nervous system drug discovery programs require the successful collection of clean CSF samples to assess exposure levels as a consequence of penetration and distribution of new chemical entities through the BBB. Rodents are the most frequently used animal model for these studies. However, collection of clean CSF samples (void of blood) has historically required terminal surgery (sampling) under anesthesia, sacrificing animals to collect a single sample. Furthermore, since only one sample could be collected per animal, the robustness of the data was rate-limiting and warranted a 3Rs alternative approach. Our goal was to refine rat CSF collection, reducing the number of animals used, and improving the quality of the samples collected in a conscious animal. We evaluated a novel indwelling Cisterna Magna (CM) catheter for ease of use, duration of use, impact on animal health, and quality of samples. This novel CM catheter reliably enabled repeated collections of CSF from a single animal for the duration of our study. Three animals were used in this validation. We were able to collect clean samples from each animal for up to 2 weeks post implantation. In addition, we were able to pair the catheter with a specialized collection device to tightly control the exact volume of CSF collected, protecting the animal's health while eliminating any samples containing blood.

BACKGROUND

The ability to collect CSF from conscious rats at multiple time points during pharmacokinetic, pharmacology, and toxicology studies, without obviating the 3Rs principles, remains a gap.

METHODS

- For high-quality rat CSF sample collection, CM cannula (RCMC-7.6, SAI Infusions Technologies) was used.
- CM cannulated rats (n=3) were received from Charles River Labs (CRL), Raleigh, NC at 6 days post implantation.
- Cannula placement is shown in Figure 1.
- The attached polyurethane tubing was exteriorized through the skin between the nape and the scapular space.
- Animals were acclimated to the facility for 24 hours post arrival.
- One person restrained the animal, one person performed the collection [see Figure 2].

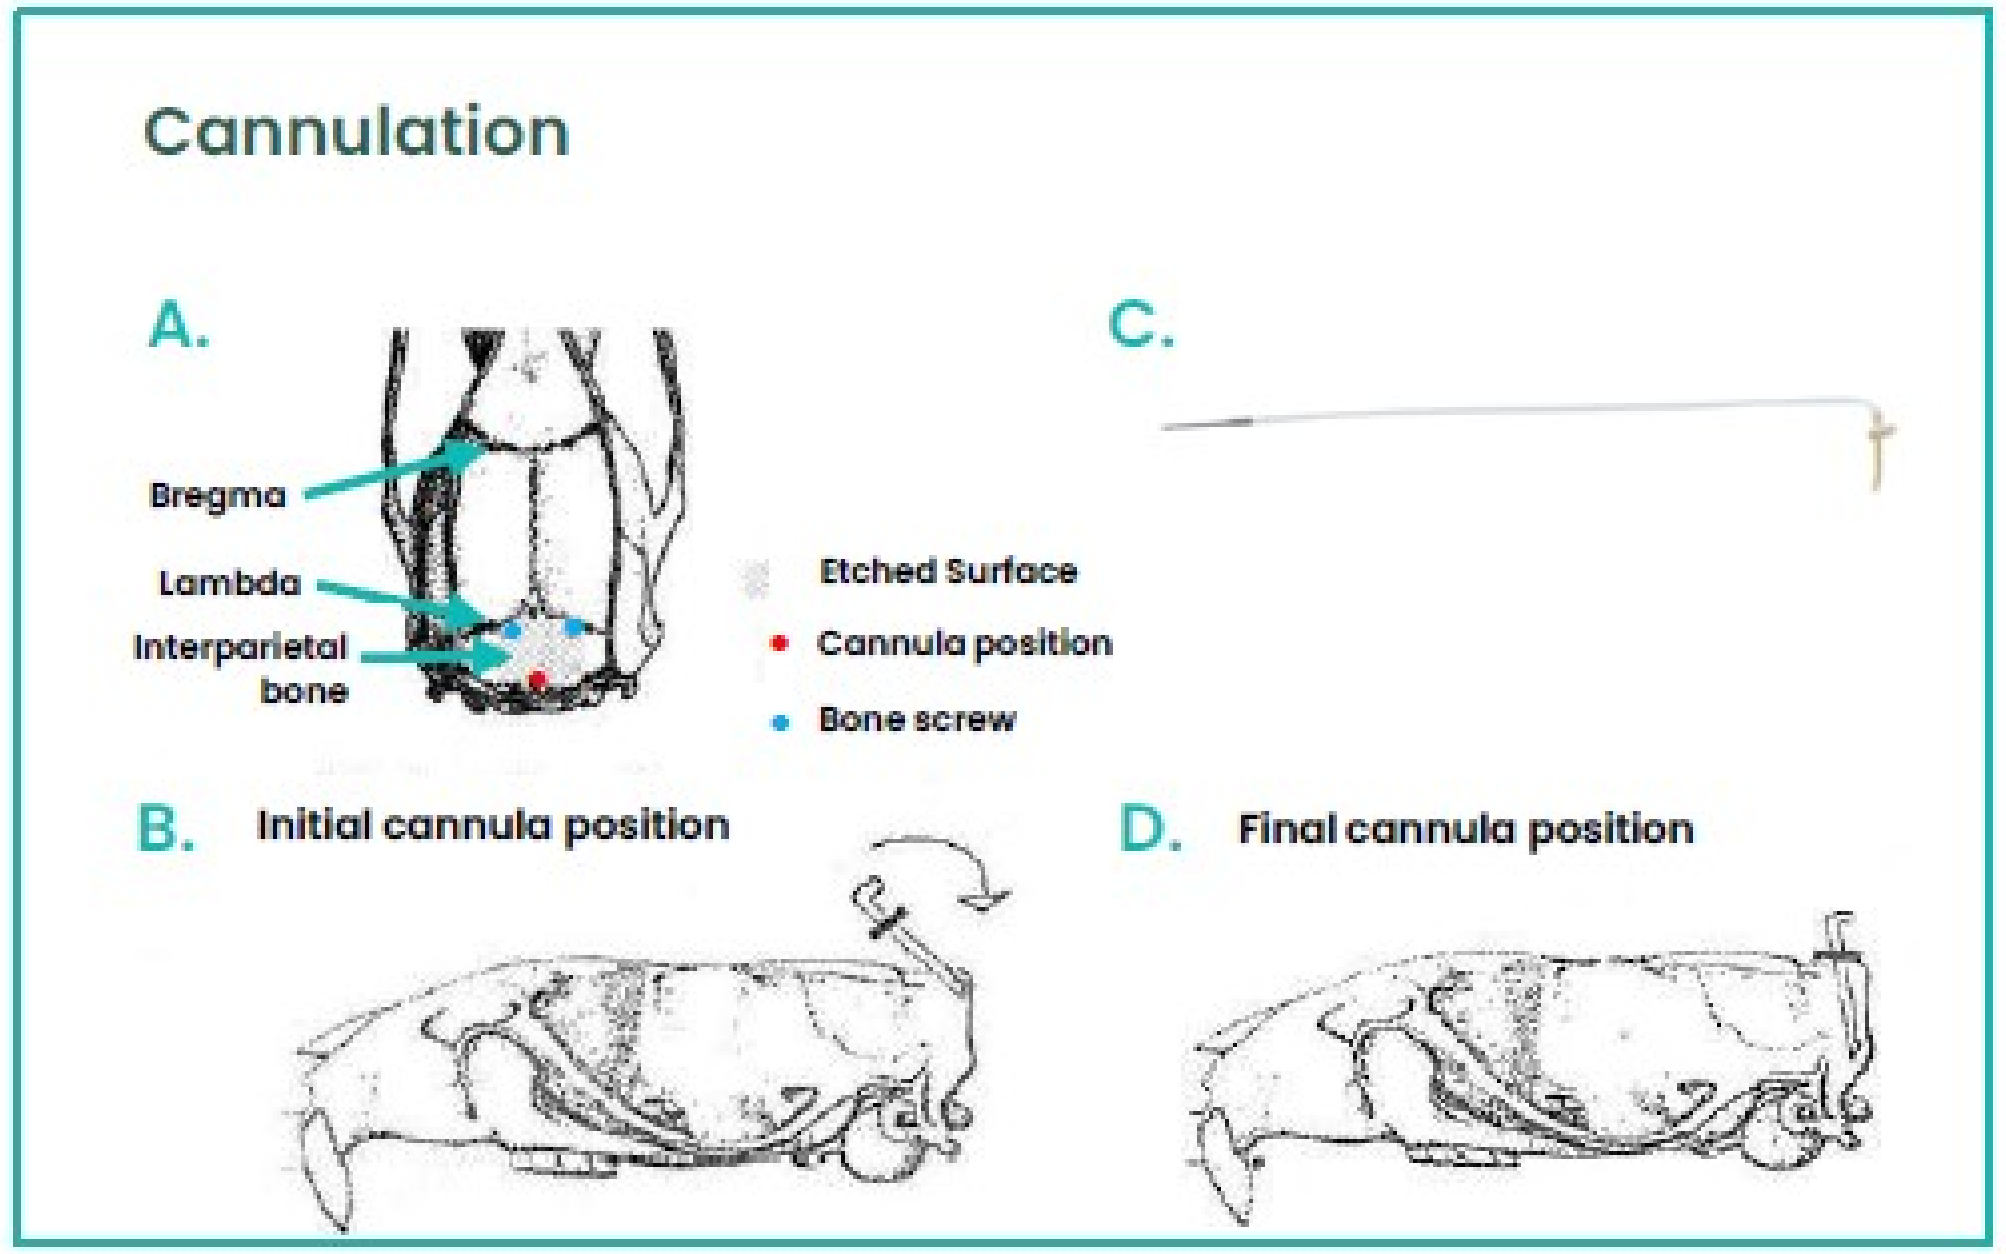
Cannulation
A.
Bregma
Lambda
Interparietal bone
Etched Surface
Cannula position
Bone screw
C.
B. Initial cannula position
D. Final cannula position

Figure 1. Rat CM Cannulation: A. skull landmarks, B. initial cannula placement, C. cannula, and D. final cannula placement

RESULTS

- Microvolume Capillary Blood Collection Tube & Plunger device (POCT-N-20; SAI Infusions Technologies) was used to ensure CSF volume collection (10, 20, 50, 100 or 200uL) and easily transferred to a cluster tube or multi-well plate for analysis.
- CSF was sampled every day/every other day until patency failure.
- At day 15 post surgery, all rats appeared healthy: 1 animal had easy, clear flow; 1 animal had a hazy sample (indicating potential infection), 1 animal had slow flow and needed syringe assistance to collect sample (still clear).
- The study was terminated at day 15 post surgery.

Collection Procedure

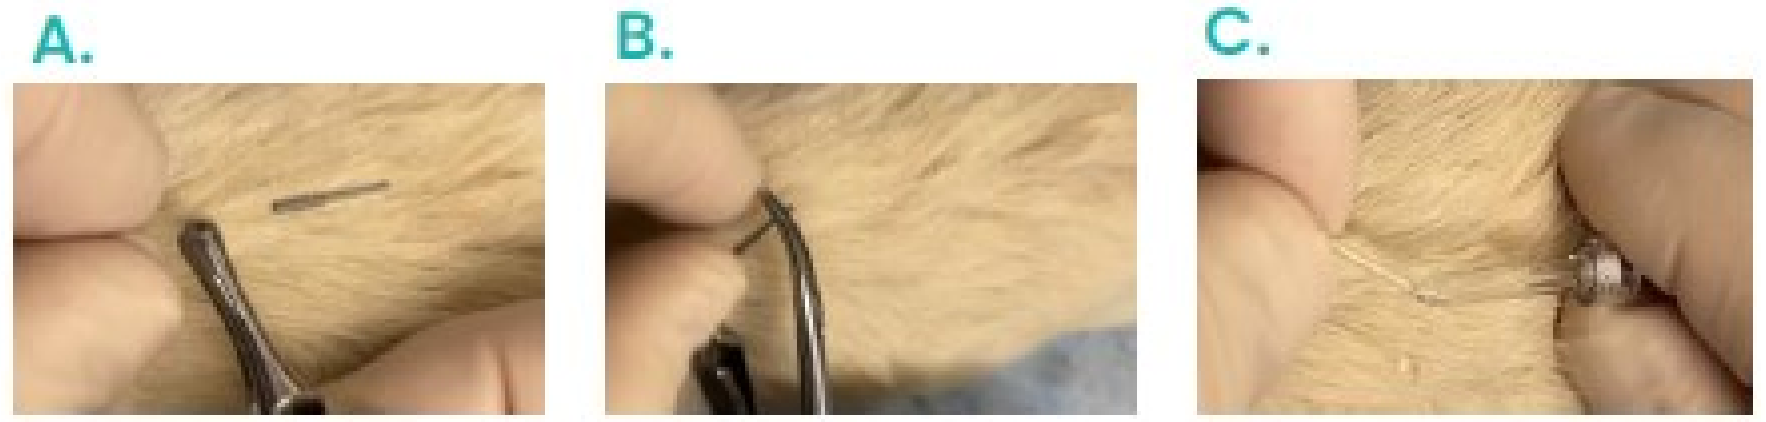
A.
B.
C.

Figure 2. Collection Procedure: A. clamp off tubing, B. remove blocker then clamp, C. collect sample vial capillary action, D. microvolume capillary tube.

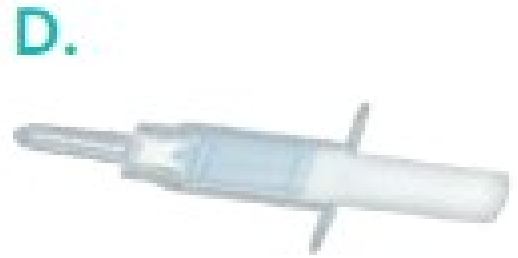
D.

CONCLUSIONS

- This new method allowed for serial, low volume (i.e., 20uL) sampling of high-quality CSF in conscious rats for up to 2 weeks post-surgical implantation.
- The procedure could allow for a reduced number of animals when collecting CSF from rats in preclinical studies.

NEUROCRINE BIOSCIENCES